First-In-Human, Multicenter Study of SENTI-202, a CD33/FLT3 Selective Off-the-Shelf Logic Gated CAR NK Cell Therapy in Hematologic Malignancies including AML: Correlative Data

Muharrem Muftuoglu[1*], Enping Hong[2*], Stephen A. Strickland[3], Alireza Eghtedar[4], Gary Schiller[5], Nosha Farhadfar[6], Ashish R. Bajel[7], Farhad Ravandi[1], Mahesh Basyal[1], Li Li[1], Lawrence Naitmazi[2], Rochelle Emery[2], Brian S. Garrison[2], Timothy Lu[2], Kanya Rajangam[2], Michael Andreeff[1]

* Denotes co-authorship. [1]The University of Texas M.D. Anderson Cancer Center, Houston, TX, [2]Senti Biosciences, Inc, South San Francisco, CA, [3]SCRI at TriStar Centennial, Nashville, TN, [4]Colorado Blood Cancer Institute, Denver, CO, [5]David Geffen School of Medicine at UCLA, Los Angeles, CA, [6]Sarah Cannon Transplant and Cellular Therapy Program at Methodist Hospital San Antonio, TX, [7]Peter MacCallum Cancer Centre, Melbourne, Australia

ABSTRACT
CT143

AACR 2025

BACKGROUND

SENTI-202 is being evaluated after lymphodepletion in patients with R/R AML in an ongoing Phase I clinical trial, SENTI-202-101 (NCT06325748). SENTI-202 is a Logic Gated CAR NK cell therapy designed to target CD33 and/or FLT3 positive hematologic malignancies including acute myeloid leukemia (AML). To achieve deeper & longer remissions, SENTI-202 was designed to also target leukemic stem cells (LSCs), which are often FLT3+ (+/-CD33+), while enabling long-term multilineage hematopoiesis by maintaining a healthy HSPC population. The SENTI-202 Logic Gated Gene Circuit therefore includes an inhibitory CAR recognizing EMCN (expressed by healthy HSPCs, rarely by AML) to protect HSPCs from potential SENTI-202 mediated off-tumor on-target toxicity even if they express CD33 and/or FLT3 & enable post-treatment hematopoietic recovery. Here, we report preliminary correlative data evaluating hematopoietic cell fractions in peripheral blood (PB) & bone marrow (BM) from 9 treated relapsed/refractory (R/R) AML patients with available data from the ongoing study (April 9th, 2025). Efficacy and clinical results were presented at AACR abstract # CT014 (oral minisymposium # CTMS01).

SENTI-202 is a First-in-Class Off-the-Shelf Logic-Gated Selective CD33 OR FLT3 NOT EMCN CAR NK Cell Therapy for Blood Cancers

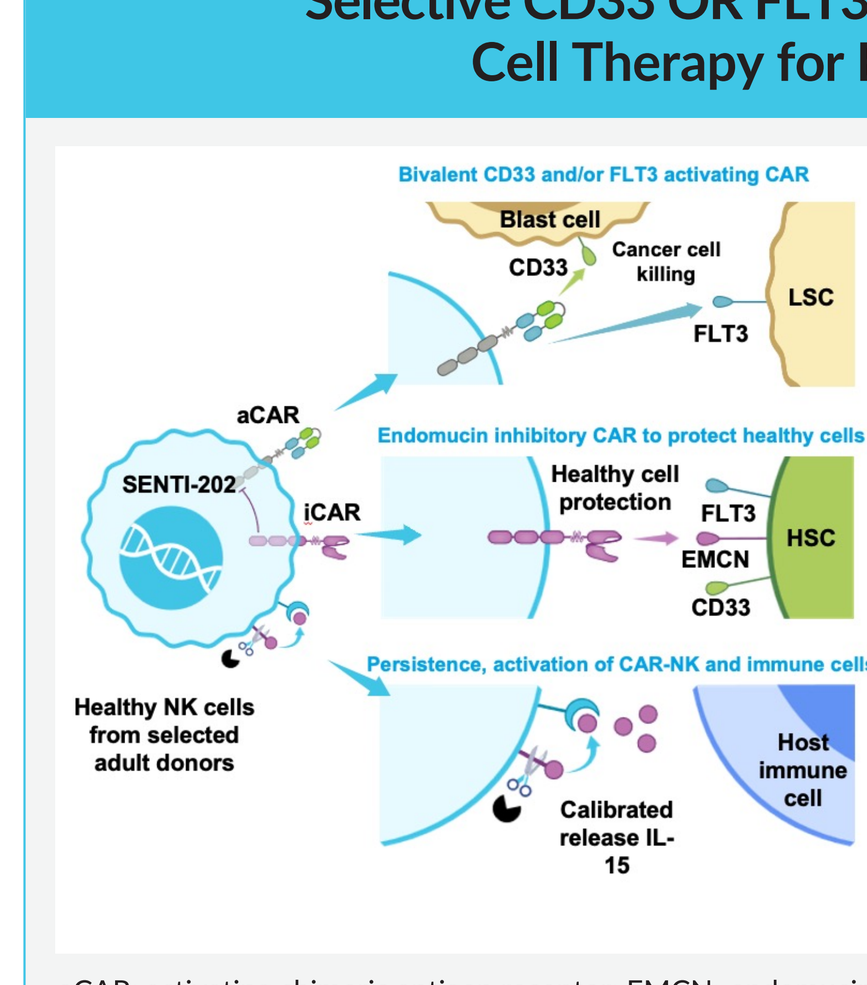
Bivalent CD33 and/or FLT3 activating CAR
Blast cell
CD33
Cancer cell killing
LSC
FLT3
aCAR
SENTI-202
iCAR
Endomucin inhibitory CAR to protect healthy cells
Healthy cell protection
FLT3
EMCN
CD33
HSC
Persistence, activation of CAR-NK and immune cells
Healthy NK cells from selected adult donors
Calibrated release IL-15
Host immune cell

SENTI-202 Gene Circuit Design

- **OR Logic Gate "Kills"** leukemia blasts and LSCs via CD33/FLT3 CAR
 - CD33 and/or FLT3 expressed in ~95% of AML patients with CD33 being predominantly expressed on bulk blasts and FLT3 on LSCs
- **NOT Logic Gate "Protects"** healthy HSC/HSPC from 'off-tumor, on-target' effects
 - Protection of HSC/HSPC via Endomucin (EMCN) inhibitory CAR even when they express CD33 and/or FLT3
 - EMCN found predominantly on healthy HSC/HSPC surface, rarely on AML blasts
- **Calibrated release IL-15 "Enhances"** SENTI-202 and host immune cell activity and persistence

aCAR: activating chimeric antigen receptor; EMCN: endomucin; HSC: hematopoietic stem cell; HSPC: hematopoietic stem and progenitor cell; iCAR: inhibitory chimeric antigen receptor; LSC: leukemic stem cell.

SENTI-202-101 Study Design

Clinical data presented at Clinical Trials Minisymposium (CTMS01, abstract CT014); **preliminary RP2D identified as Schedule I, Dose Level 2**

Patient Population
- Adult patients
- R/R CD33 and/or FLT3 expressing heme malignancies including AML/MDS
- Received 1-3[2] prior AML treatments including targeted agents if applicable

Study Design
- "3+3" study design
- Dose escalation followed by disease-specific expansion cohorts AML and MDS
- Two dose levels, two dose schedules[3]
- Plans to transition from Phase 1 to pivotal study

Key Endpoints
- Safety, DLT, identify RP2D
- Efficacy, including bone marrow recovery and MRD
- Pharmacokinetics, pharmacodynamics, CyTOF

Multi-Dose Cycle

SENTI-202 Dose: Dose Level	CAR+ NK Cells/Dose
1	1 x 10^9
2	1.5 x 10^9

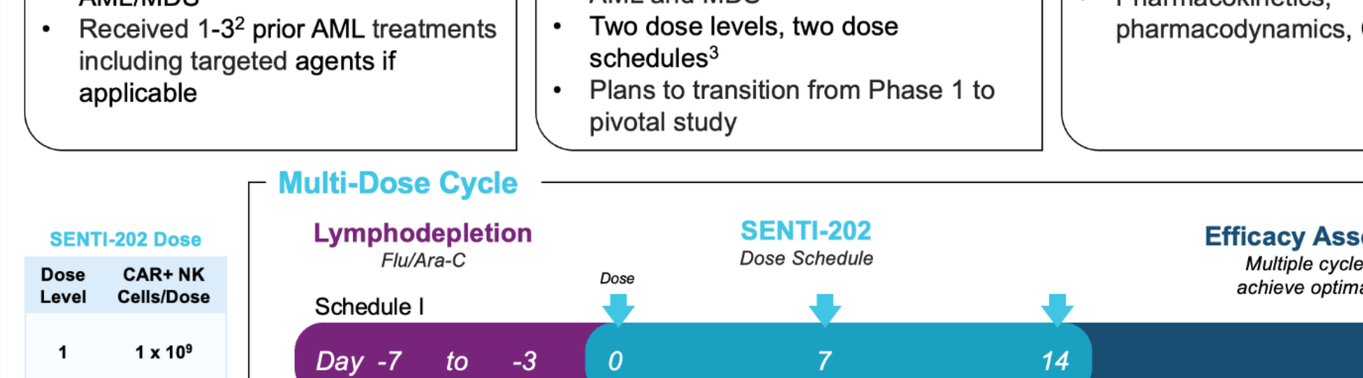
Lymphodepletion
Flu/Ara-C
SENTI-202
Dose Schedule
Efficacy Assessment
Multiple cycles allowed to achieve optimal response[4]
Dose
Schedule I
Day -7 to -3
0
7
14
28
Schedule II
Day -7 to -3
0
3
7
10
14
28

DLT: dose limiting toxicity; MRD: measurable residual disease; R/R: relapsed refractory; RP2D: recommended phase 2 dose
[1]NCT06325748; [2]1-2 prior for MDS; [3]Other dose levels and schedules may be evaluated based on study data;
[4]Subjects in MRD negative complete remission may receive one additional cycle as consolidation.

SENTI-202-101 Dosing and Correlative Sampling Timepoints

Methods

- Whole blood and bone marrow aspirate are collected into sodium heparin tubes, then processed to cryopreserved PBMC/BMMC by Ficoll separation.
- Pharmacokinetics was determined by detection of the SENTI-202 transgene in PBMC samples by droplet digital PCR.
- BMMC samples were analyzed by a 49-marker mass cytometry (CyTOF) panel.
- Additional whole blood samples were collected into Cyto-Chex BCT tubes and analyzed by flow cytometry.

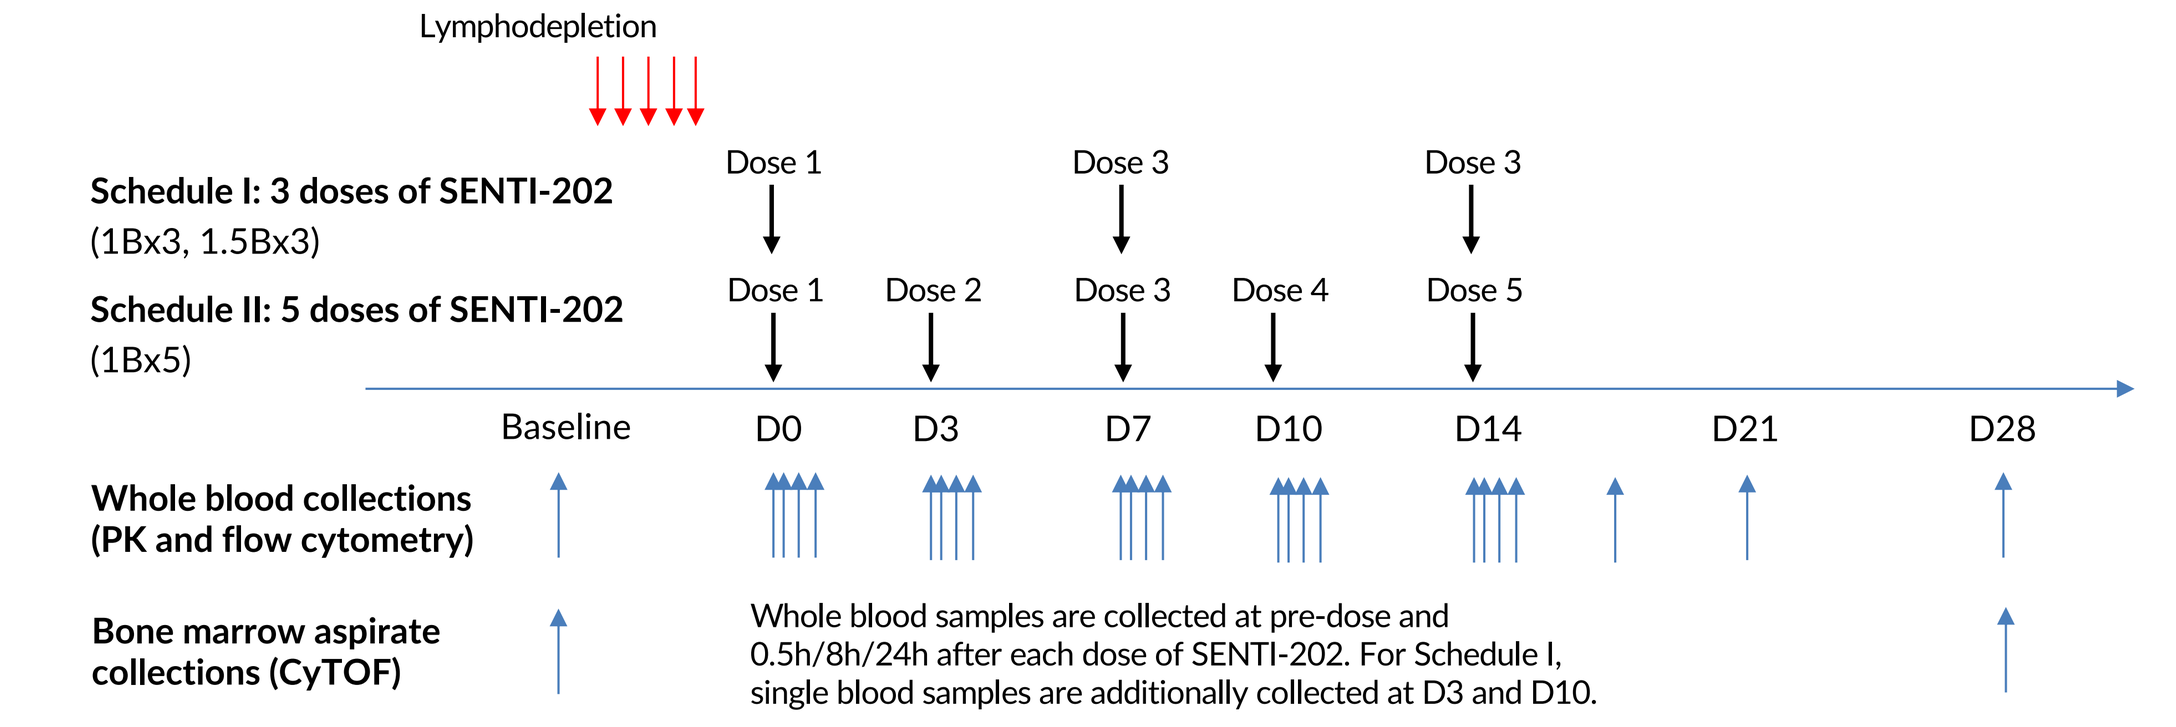
Lymphodepletion
Schedule I: 3 doses of SENTI-202
(1Bx3, 1.5Bx3)
Dose 1
Dose 3
Dose 3
Schedule II: 5 doses of SENTI-202
(1Bx5)
Dose 1
Dose 2
Dose 3
Dose 4
Dose 5
Baseline
D0
D3
D7
D10
D14
D21
D28
Whole blood collections
(PK and flow cytometry)
Bone marrow aspirate collections (CyTOF)
Whole blood samples are collected at pre-dose and 0.5h/8h/24h after each dose of SENTI-202. For Schedule I, single blood samples are additionally collected at D3 and D10.

SENTI-202 Pharmacokinetics: SENTI-202 is Detected in All Treated Patients

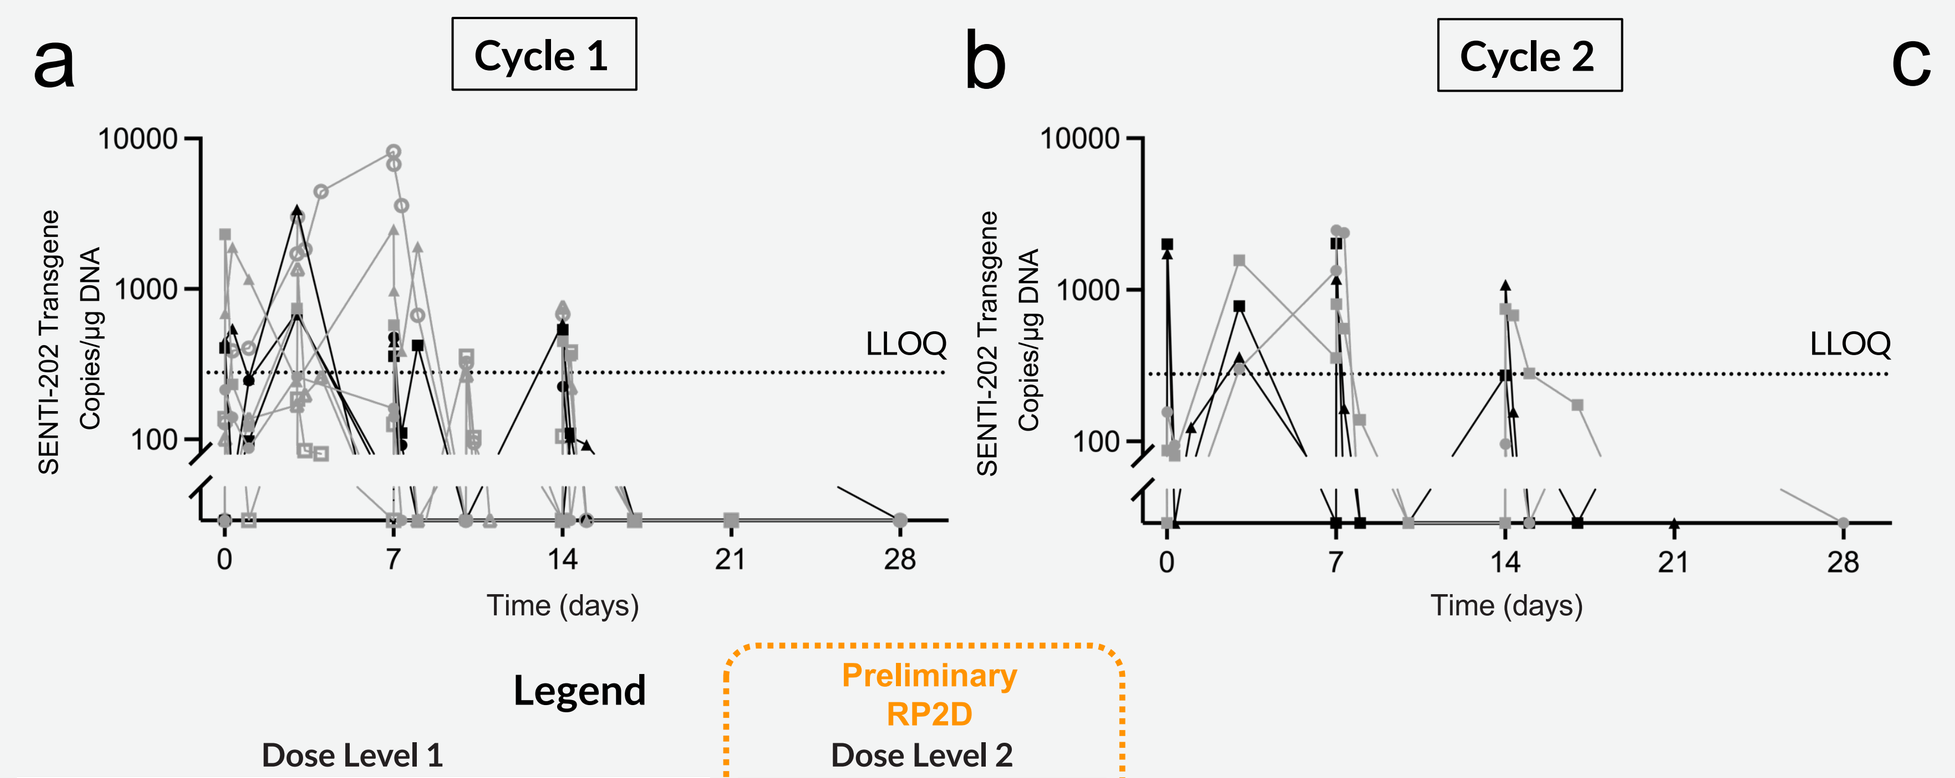
a
Cycle 1
b
Cycle 2
SENTI-202 Transgene Copies/μg DNA
10000
1000
100
LLOQ
0
7
14
21
28
Time (days)
Legend
Dose Level 1
Schedule I
Subject 1 (CR MRD-)
Subject 2 (CR MRD-)
Subject 3 (PD)
Schedule II
Subject 7 (MLFS)
Subject 8 (TBD)
Subject 9 (TBD)
Preliminary RP2D
Dose Level 2
Schedule I
Subject 4 (CR MRD-)
Subject 5 (CRh MRD-)
Subject 6 (SD)

c

PK Parameters Median (range) given for all	Dose Level 1 (1B/CAR+ NK dose) Schedule I (3 doses) N=3	Dose Level 1 (1B/CAR+ NK dose) Schedule II (5 doses) N=3	Dose Level 2 (1.5B CAR+ NK/dose) Schedule I (3 doses) N=3 (Preliminary RP2D)	All Subjects N=9
C_{max} (Copies/μg DNA)	2392 (1567 - 2496)	1358 (380 - 8212)	1739 (676 - 3388)	1878 (380 - 8212)
T_{max} (Days)	5 (0 - 7)	7 (3 - 14)	3 (0 – 7)	3 (0 - 14)
AUC_{obs} (Days*Copies/μg DNA)	6873 (3160 - 11074)	1554 (720 - 29067)	3532 (2133 - 12395)	3593 (720 - 29067)
Peripheral clearance (Days)	11 (7 - 15)	14 (14 - 15)	13 (7 – 14)	14 (7 - 15)

Pharmacokinetic (PK) analysis of SENTI-202 in trial subjects. SENTI-202 was detected in all treated subjects, with data shown from (a) Cycle 1 and (b) Cycle 2. SENTI-202 PK profile was consistent with allogeneic NK cell therapy with cells detected in the first 2 weeks in periphery followed by clearance thereafter. (c) PK parameters were analyzed and showed no significant differences in exposure between Schedule I dose levels 1 and 2, or cycles 1 and 2. Estimated LLOQ assumes equivalent DNA loading for each reaction. Note: Subject 1 Cycle 1 samples were processed out of stability window and are excluded from analysis of PK parameters.

SUBJECT BASELINE AML MARKER EXPRESSION

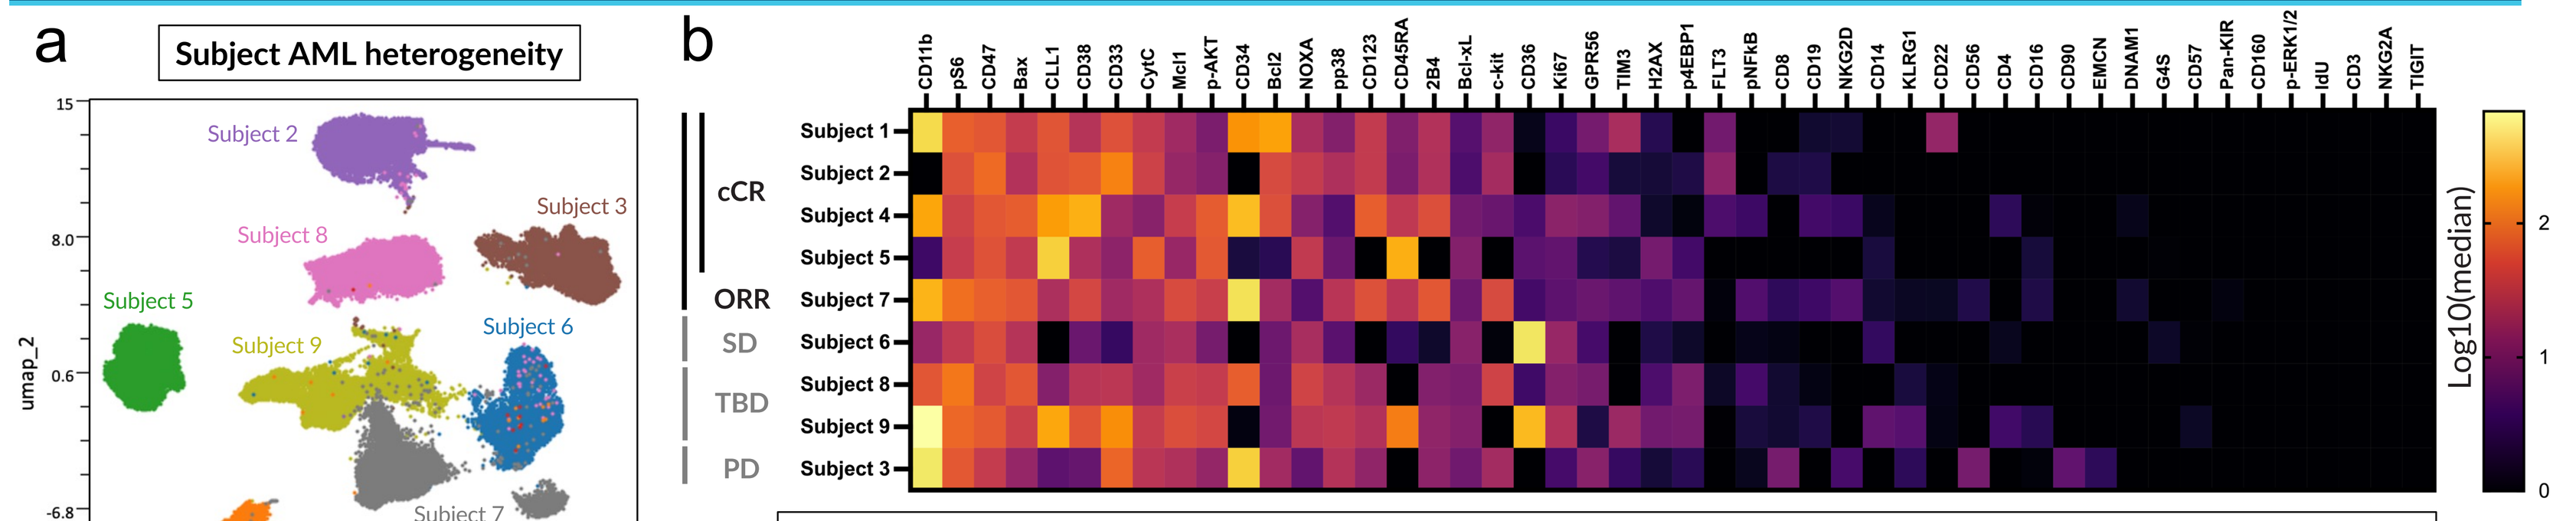
a
Subject AML heterogeneity
Subject 2
Subject 8
Subject 3
Subject 5
Subject 9
Subject 6
Subject 7
Subject 4
Subject 1
umap_1
umap_2
b
CD11b
pS6
CD47
Bax
CLL1
CD38
CD33
CytC
Mcl1
p-AKT
CD34
Bcl2
NOXA
pp38
CD123
CD45RA
2B4
Bcl-xL
c-kit
CD36
Ki67
GPR56
TIM3
H2AX
p4EBP1
FLT3
pNFkB
CD8
CD19
NKG2D
CD14
KLRG1
CD22
CD56
CD4
CD16
CD90
EMCN
DNAM1
G4S
CD57
Pan-KIR
CD160
p-ERK1/2
IdU
CD3
NKG2A
TIGIT
cCR
ORR
SD
TBD
PD
Subject 1
Subject 2
Subject 4
Subject 5
Subject 7
Subject 6
Subject 8
Subject 9
Subject 3
Log10(median)

- CD33, CLL1, and CD123 are expressed highly in all subjects who achieved cCR.
- 3 of 4 subjects who achieved cCR expressed FLT3; whereas both the SD and the no-response subject had < 3% FLT3 expression. Note: Subject 3 had a more stem/progenitor-like AML, expressing stem-associated markers (CD34+, CD90+, EMCN+) and mutant TP53, that did not respond to treatment. Subject 3 AML was also 43% EMCN+ (EMCN is rarely expressed in AML).

The SENTI-202-101 clinical trial enrolled subjects with heterogenous AML profiles. (a) UMAP analysis of AML populations in bone marrow samples at baseline show diverse proteomic features that uniquely identify samples from each subject. (b) Heat map analysis of AML populations in bone marrow samples show the prevalence of AML-associated markers and their heterogeneity of expression across study subjects. cCR, composite complete response; PD, progressive disease / no response; SD, stable disease; TBD, to be determined.

BASELINE BONE MARROW ANALYSIS

Low Expression High	Subject 1 1Bx3	Subject 2 1Bx3	Subject 4 1.5Bx3 (Prelim. RP2D)	Subject 5 1.5Bx3 (Prelim. RP2D)	Subject 7 1Bx5	Subject 6 1.5Bx3 (Prelim. RP2D)	Subject 8 1Bx5	Subject 9 1Bx5	Subject 3 1Bx3
Best Response	CR MRD-	CR MRD-	CR MRD-	CRh MRD-	MLFS	SD	TBD- continuing Rx after SD / Cycle 1	TBD- continuing Rx after SD / Cycle 1	PD
Mutations & cytogenetics at diagnosis	ASXL1, ETV6, NRAS	FLT3-ITD, TET2	IDH2mut, SRSF2, PSU DIC (2;4)(q31;q21)X2,t(13,21)(q32;q22)X2/46,XY	NRAS, TET2, SRSF2, CEBPA, ASXL1, RUNX1	SF3B1, NRAS, KRAS, GATA2, t(I;16)(q21,q22)	RPN1:MECOM, paracentric inversion of chromosome 3, GATA2, NRAS, WT1	RUNX1, NF1, 46,XY,t(4;17)(q31;q11.2)	TET2, FLT3-ITD, SRSF2, KRAS, ASXL1, RUNX1, SETBP1	loss of 3q, 5q, 7q; DNMT3A, KRAS, TP53
Mutations & cytogenetics at study entry	WT1, ASXL1, EGFR, ETV6, NF1, PFH6, gain of NUP214 (9q34), gain of ABL1 (9q34), gain of CDKN2A (9p21), trisomy 9, TRA/D rearrangement	TET2	IDH2mut, SRSF2, add(2)(q33)X2, add(7)(p15)X2, add(13)(q32)X2, +mar/46,XY, tri/tetra/polisomy	SH2B3, SRSF2, CEBPA, TET2, ASXL1, RUNX1	GATA2, SF3B1, inv(3)(q21q26.2), der(1;16)(q10;p10),	46,XY,inv(3)(q21q26.2), GATA2, NRAS, WT1	RUNX1, IKZF1, 46,XY,t(4;17)(q31;q11.2)	ASXL1, CBL, FLT3, RUNX1, SETBP1, SRSF2, TET2, del(7)(p14p12)/46	CBL, DNMT3A, KRAS, TP53, loss of 5q, 7q.
Cell populations: Baseline (AML blasts, Monocytic, Erythroid, HSC/HSPCs, CD4 T cells, CD8 T cells, NK cells, B cells)	C1D28 sample (PR) used due to low baseline sample viability.								
CD33 (baseline, disease outlined in red)									
%CD33+	Site: 40% CyTOF: 96.6%	Site: 98.8% CyTOF: 99.1%	Site: Dim/Partial CyTOF: 67.1%	Site: Positive CyTOF: 60.1%	Site: Positive CyTOF: 62.3%	Site: 88.7% CyTOF: 28.1%	Site: 98.5% CyTOF: 72.9%	Site: 78.5% CyTOF: 73.1%	Site: Positive CyTOF: 97.1%
FLT3 (baseline, disease outlined in red)									
%FLT3+	CyTOF: 23.8%	CyTOF: 45.6%	CyTOF: 5.0%	CyTOF: 0.8%	CyTOF: 0.4%	CyTOF: 2.7%	CyTOF: 2.1%	CyTOF: 1.9%	CyTOF: 0.1%

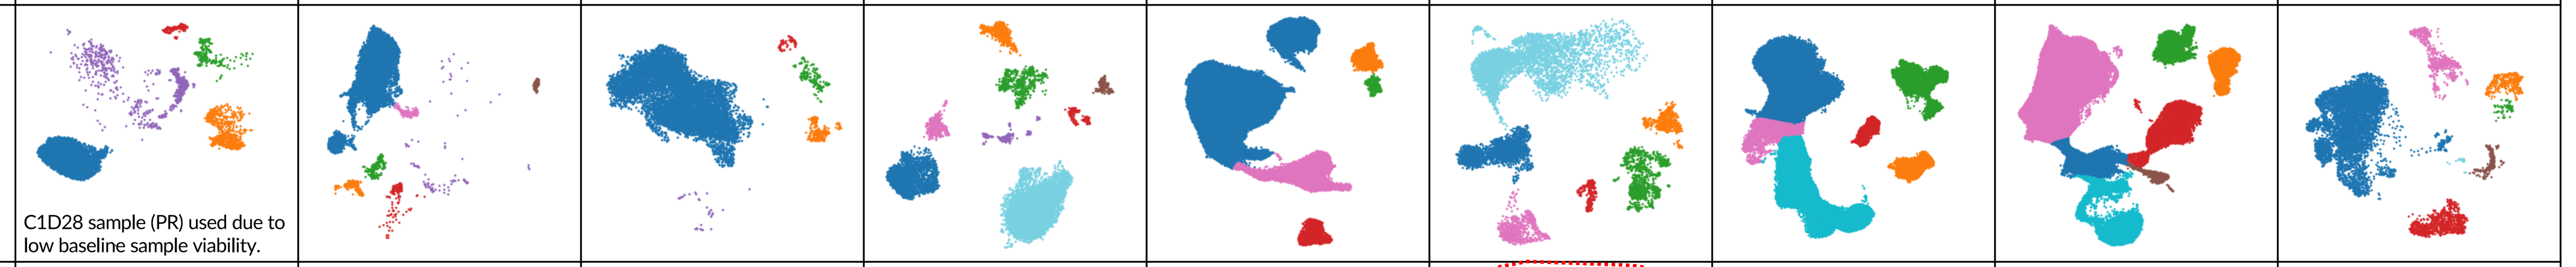

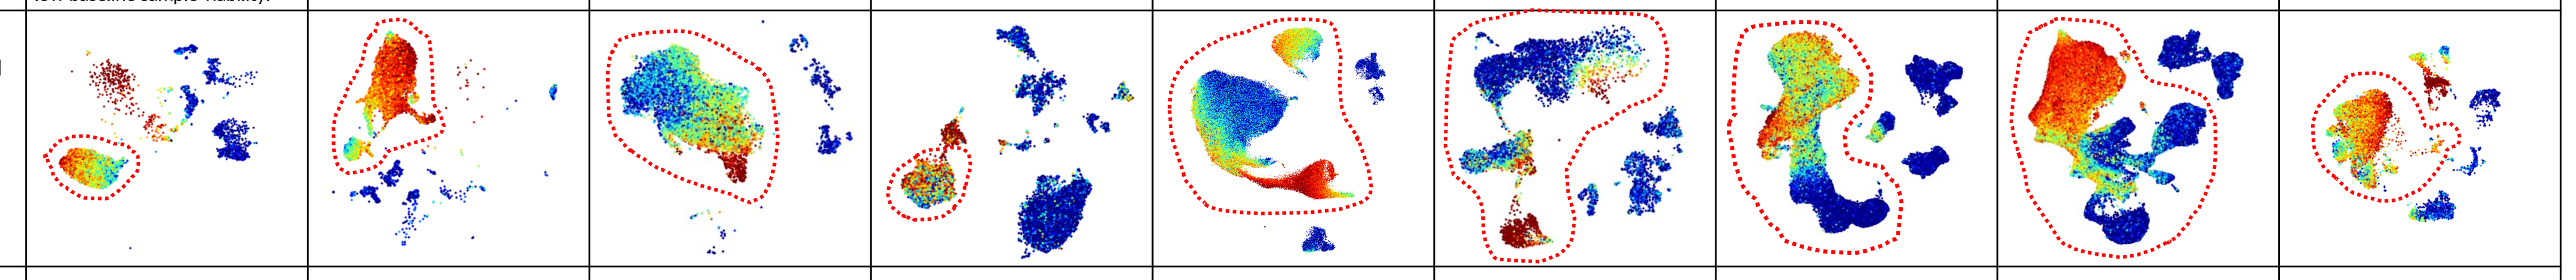

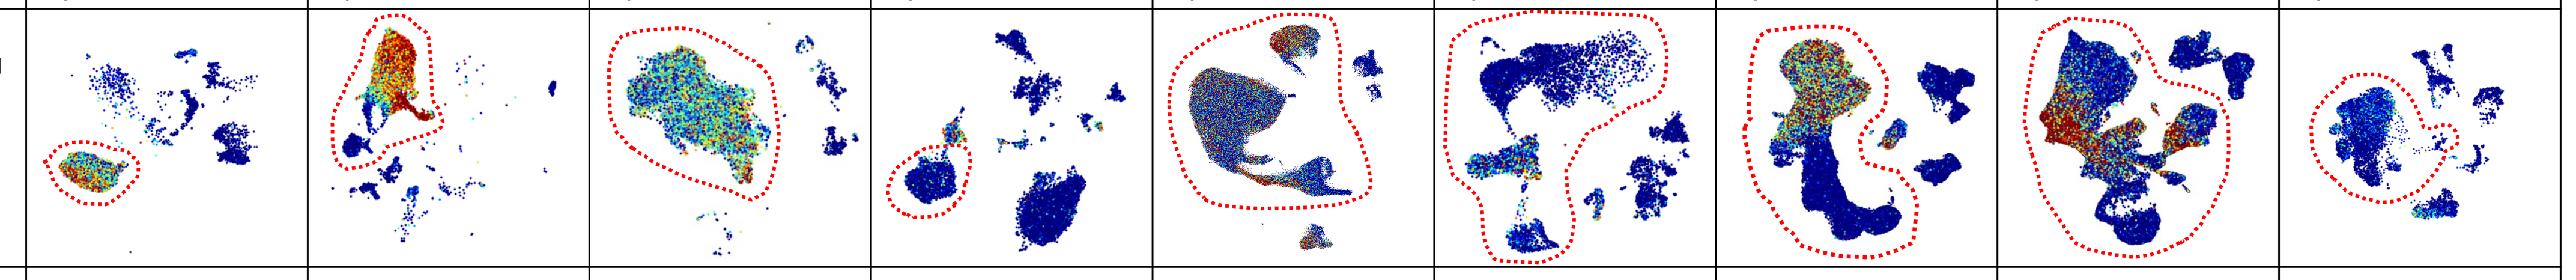

Responses After SENTI-202 Treatment

Cell populations: Best Response (end of Cycle 1 for subjects 8 & 9)

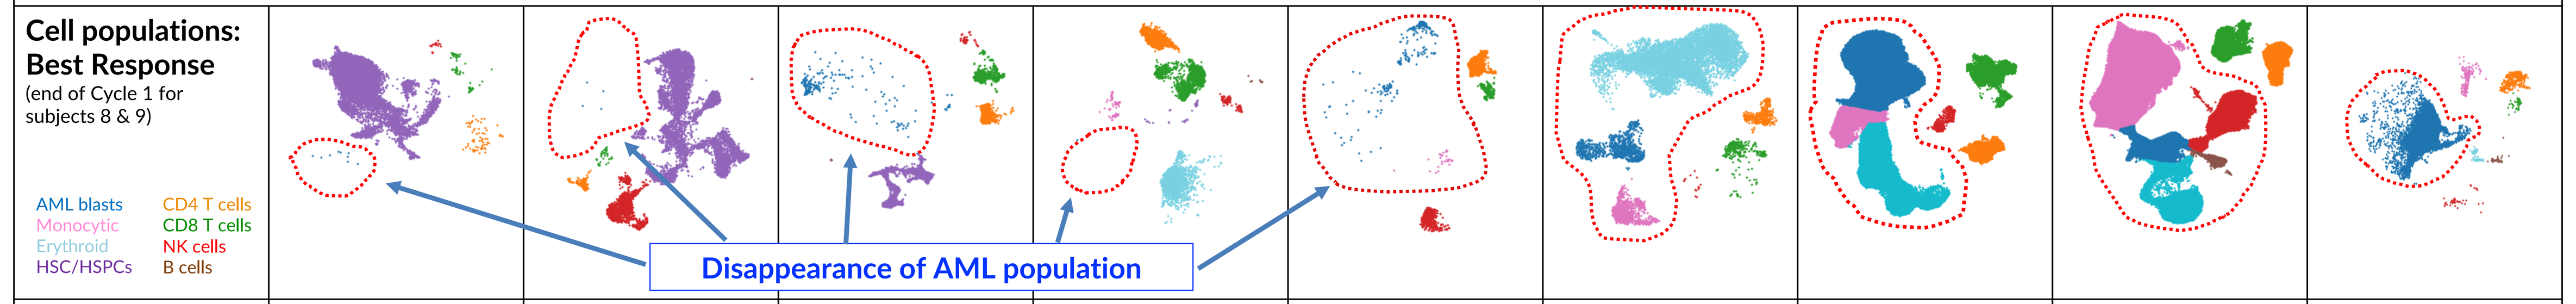
AML blasts
Monocytic
Erythroid
HSC/HSPCs
CD4 T cells
CD8 T cells
NK cells
B cells
Disappearance of AML population

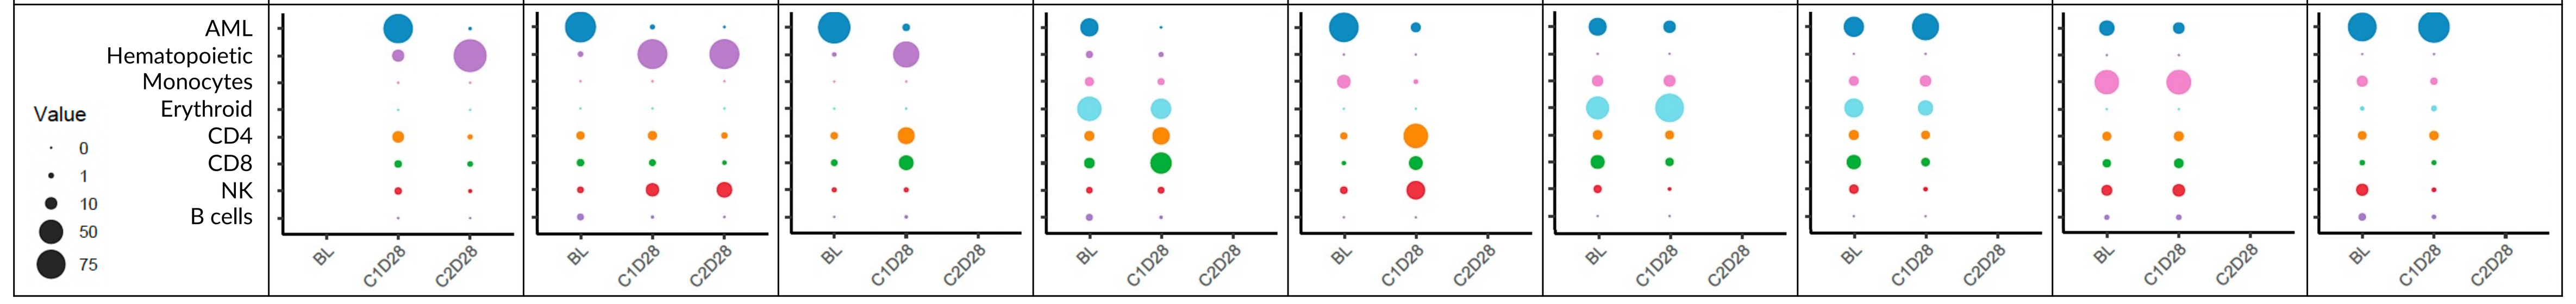
AML
Hematopoietic
Monocytes
Erythroid
CD4
CD8
NK
B cells
Value
0
1
10
50
75
BL
C1D28
C2D28

BLAST & LSC REDUCTION IN cCR BONE MARROW

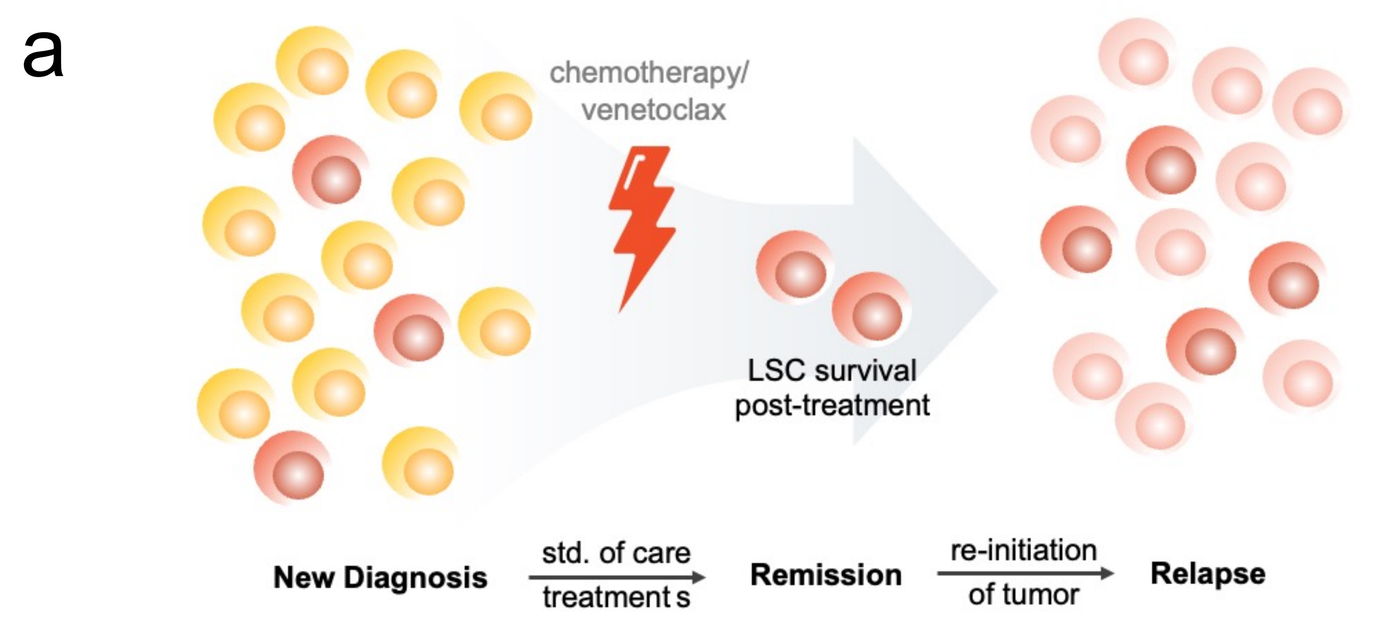
a
chemotherapy/ venetoclax
LSC survival post-treatment
New Diagnosis
std. of care treatments
Remission
re-initiation of tumor
Relapse
AML blast
AML LSC
AML blast (relapse)

b

LSCs at baseline are largely non-cycling and not expected to be eliminated by lymphodepleting chemotherapy.

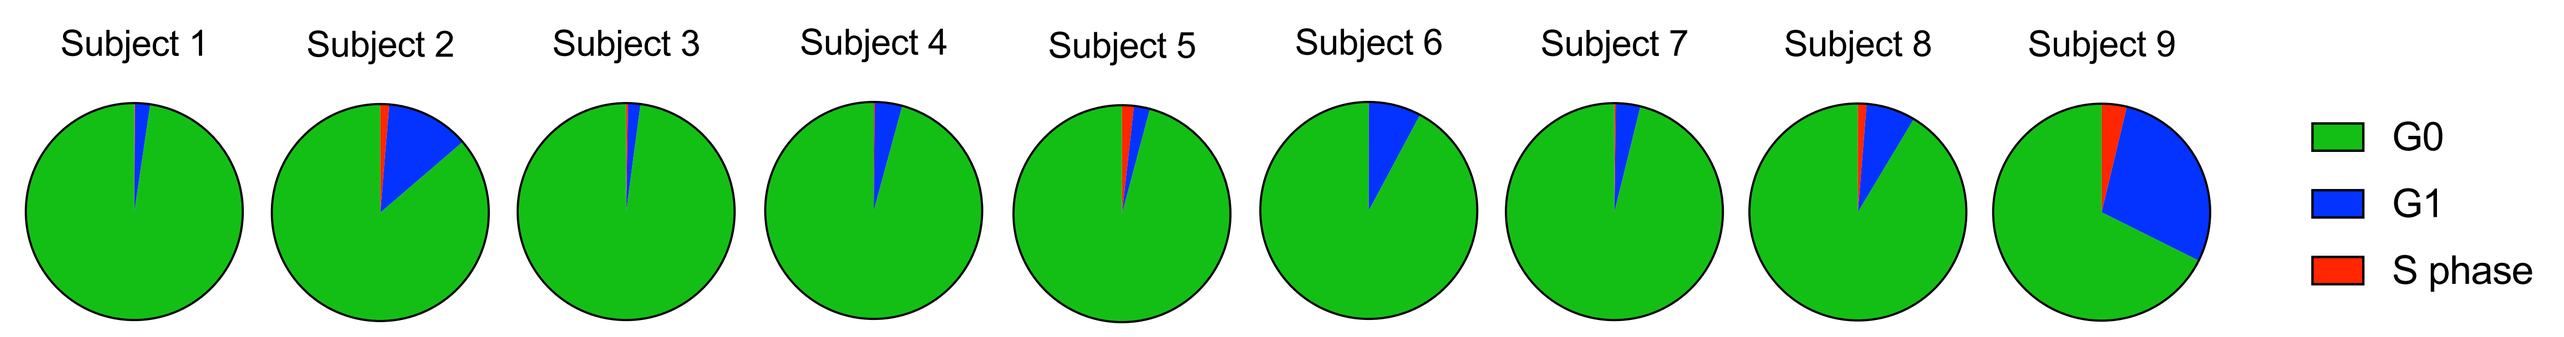
Subject 1
Subject 2
Subject 3
Subject 4
Subject 5
Subject 6
Subject 7
Subject 8
Subject 9
G0
G1
S phase

c

CD33 & FLT3 expression in LSCs

	Subject	Best Response	%CD33+	%FLT3+	%CD33+ FLT3+
Preliminary RP2D	Subject 4	cCR	93.2	82.1	76.8
Preliminary RP2D	Subject 5	cCR	81.9	6.6	5.4
Preliminary RP2D	Subject 6	SD	72.7	40.5	30.7
	Subject 1	cCR	98.6	89.5	88.6
	Subject 2	cCR	95.4	62.9	62.6
	Subject 3	PD	99.5	27.8	27.8
	Subject 7	MLFS	61.3	39.0	24.1
	Subject 8	TBD	97.6	69.5	68.0
	Subject 9	TBD	70.4	23.2	18.4

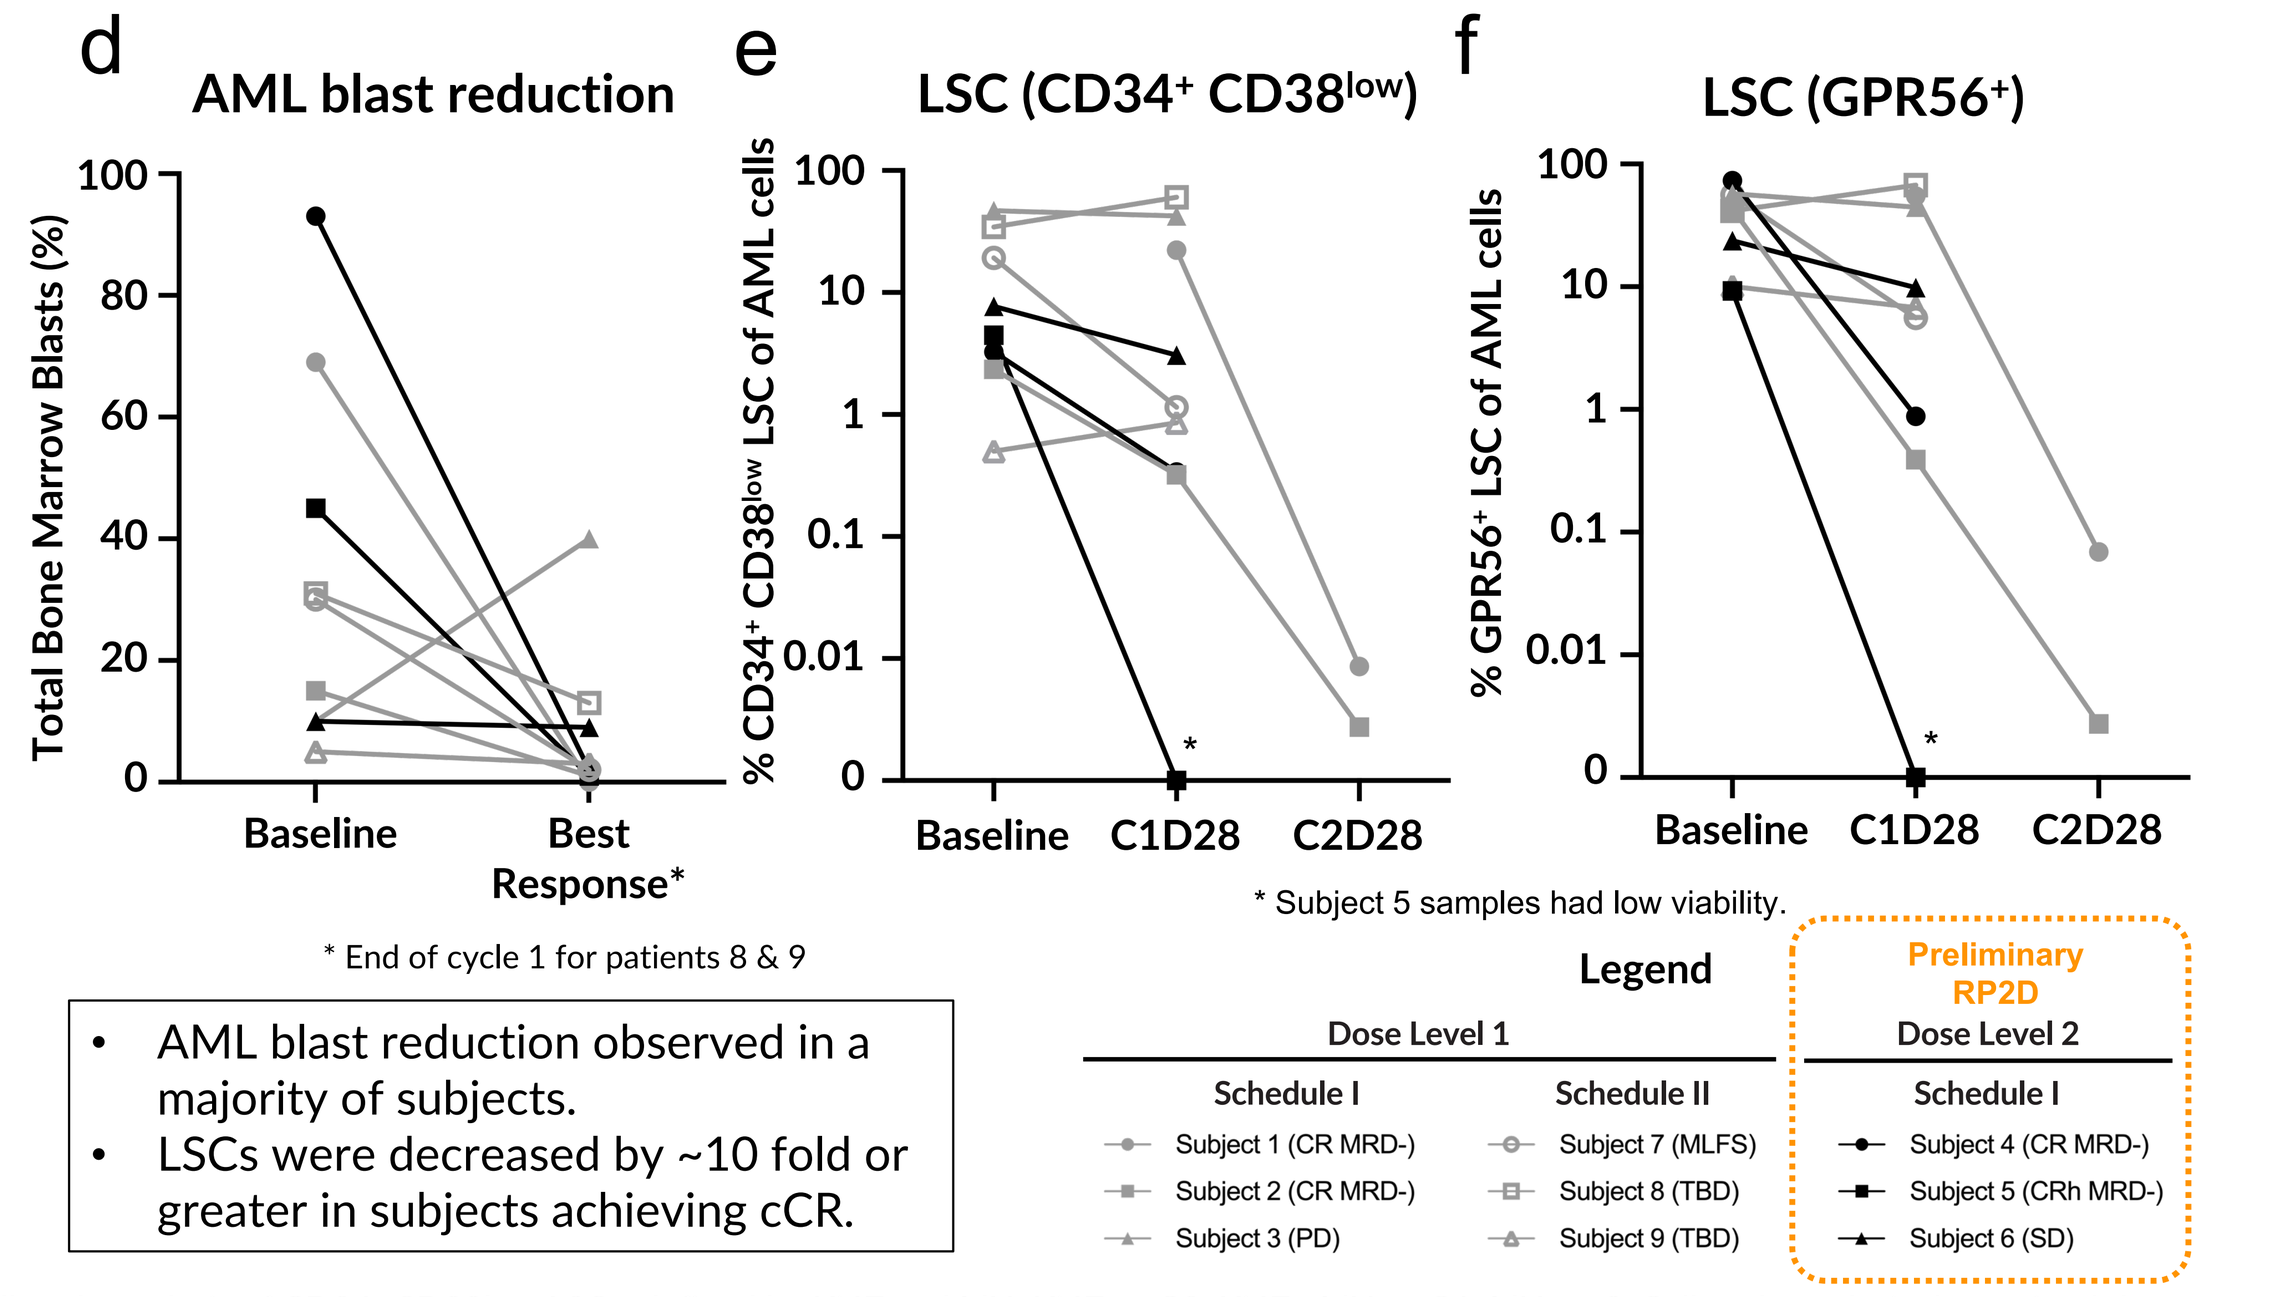
d
AML blast reduction
Total Bone Marrow Blasts (%)
Baseline
Best Response*
* End of cycle 1 for patients 8 & 9
e
LSC (CD34+ CD38low)
% CD34+ CD38low LSC of AML cells
f
LSC (GPR56+)
% GPR56+ LSC of AML cells
Baseline
C1D28
C2D28
* Subject 5 samples had low viability.
Legend
Dose Level 1
Schedule I
Subject 1 (CR MRD-)
Subject 2 (CR MRD-)
Subject 3 (PD)
Schedule II
Subject 7 (MLFS)
Subject 8 (TBD)
Subject 9 (TBD)
Preliminary RP2D
Dose Level 2
Schedule I
Subject 4 (CR MRD-)
Subject 5 (CRh MRD-)
Subject 6 (SD)

- AML blast reduction observed in a majority of subjects.
- LSCs were decreased by ~10 fold or greater in subjects achieving cCR.

Analysis of leukemic stem cells (LSCs) in trial subjects. (a) Schematic of clonal repopulation of LSCs after treatment with conventional anti-AML chemotherapy. (b) Cell cycle analysis of CD34+CD38low LSCs, defined by Ki67 and IdU: Ki67- = G0, Ki67+ IdU- = G1, IdU+ = S. Cell cycle analysis determined that most LSCs in treated subjects were quiescent at baseline, and therefore not expected to be eliminated by lymphodepleting chemotherapy such as fludarabine/ cytarabine. (c) LSCs identified in subject bone marrow were shown to express CD33 and FLT3, and can therefore be targeted by SENTI-202. (d) AML blast reduction (measured locally by flow cytometry) was noted in a majority of subjects across all dosing cohorts. LSCs in responders were identified by two different methods: (e) CD34+CD38low and (f) GPR56+, and their proportions in subject bone marrow were shown to decrease at least 10-fold after SENTI-202 treatment. * Subject 5 samples in (e) and (f) had low viability.

HSC RETENTION AND IMMUNE REPOPULATION IN cCR SUBJECTS

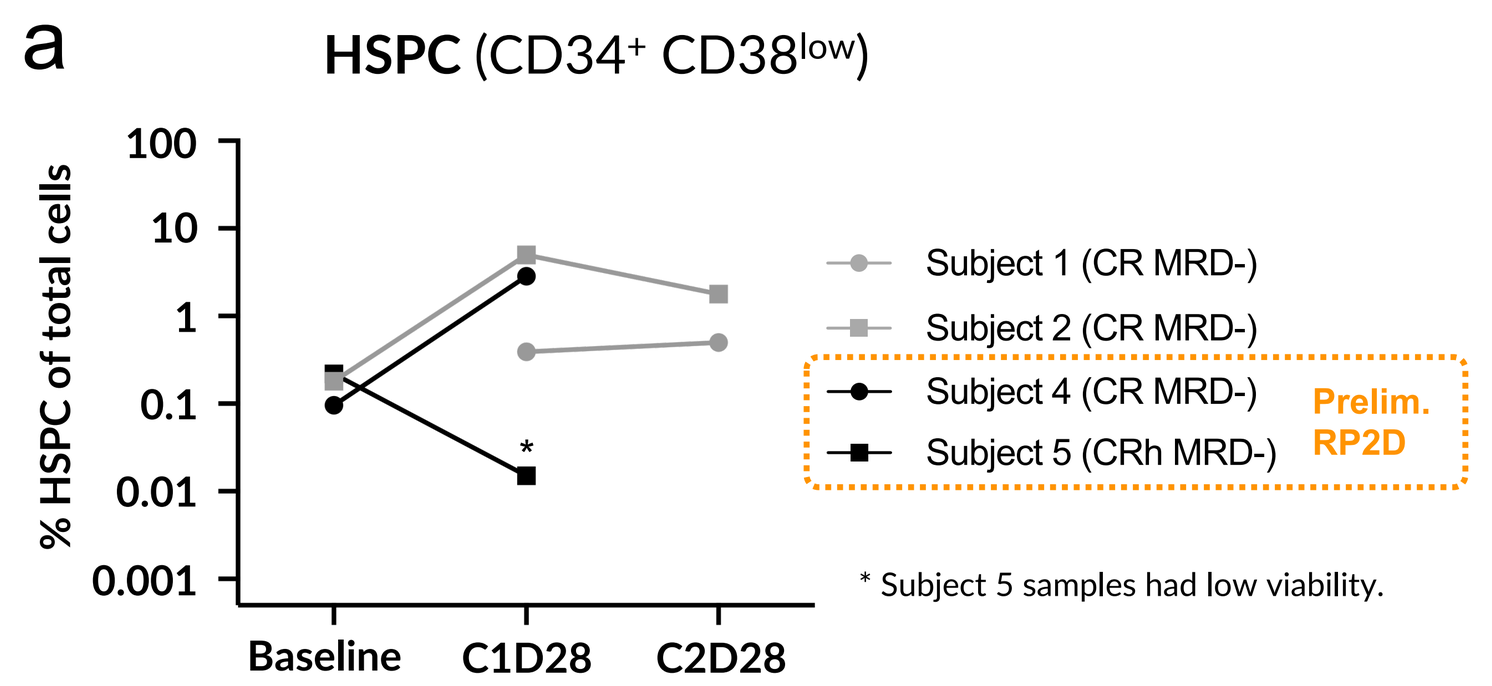
a
HSPC (CD34+ CD38low)
% HSPC of total cells
Baseline
C1D28
C2D28
Subject 1 (CR MRD-)
Subject 2 (CR MRD-)
Subject 4 (CR MRD-)
Subject 5 (CRh MRD-)
Prelim. RP2D
* Subject 5 samples had low viability.

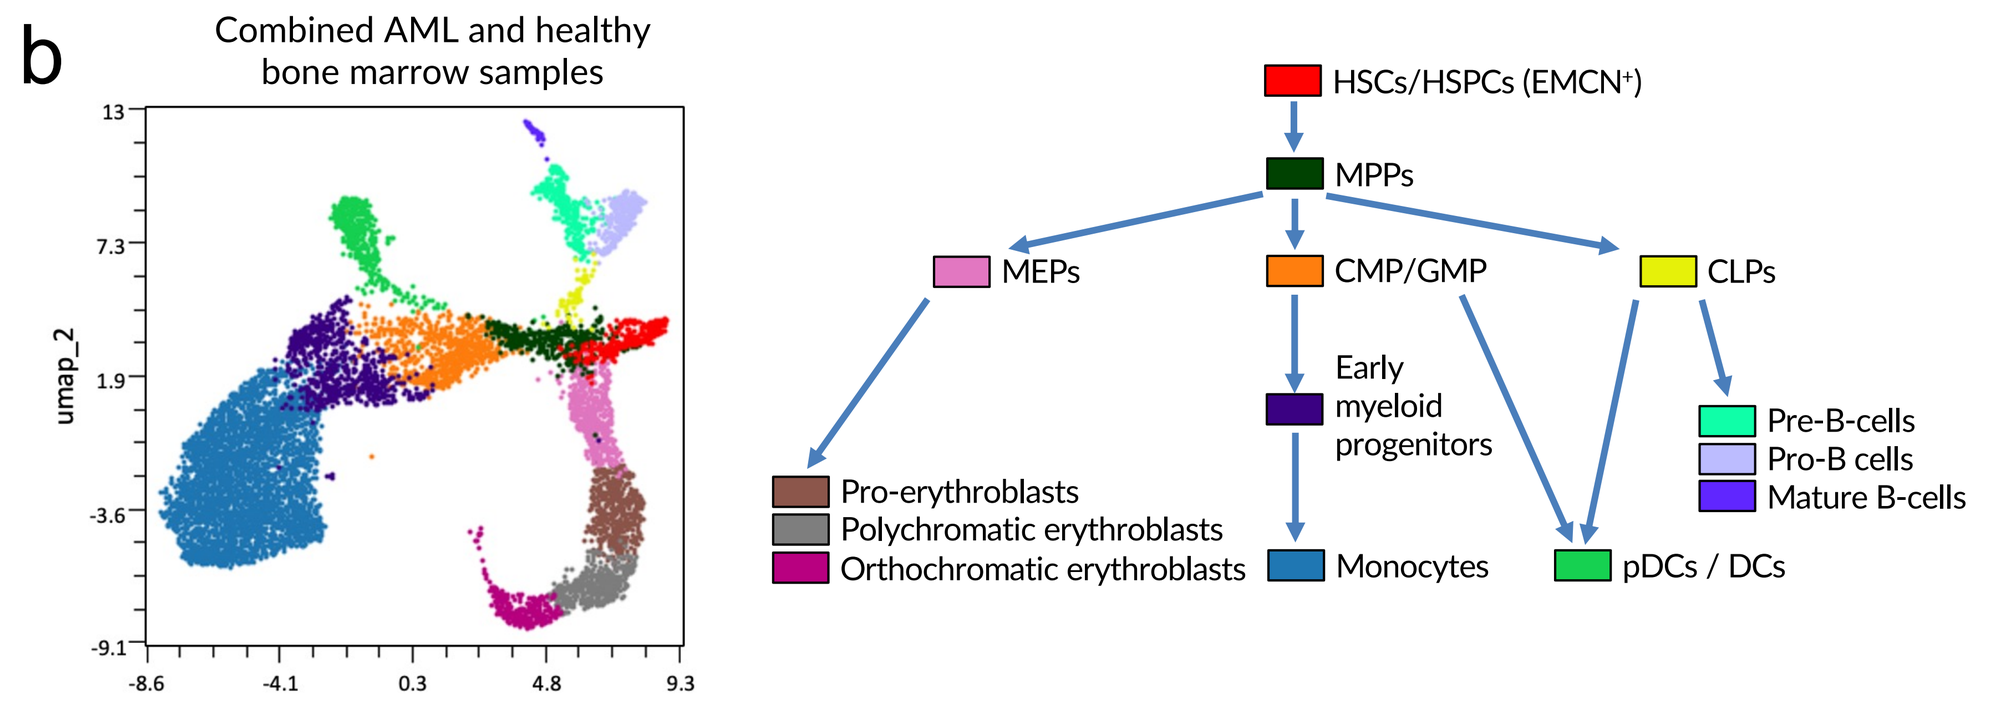
b
Combined AML and healthy bone marrow samples
umap_1
umap_2
HSCs/HSPCs (EMCN+)
MPPs
MEPs
CMP/GMP
CLPs
Early myeloid progenitors
Pro-erythroblasts
Polychromatic erythroblasts
Orthochromatic erythroblasts
Monocytes
pDCs / DCs
Pre-B-cells
Pro-B cells
Mature B-cells

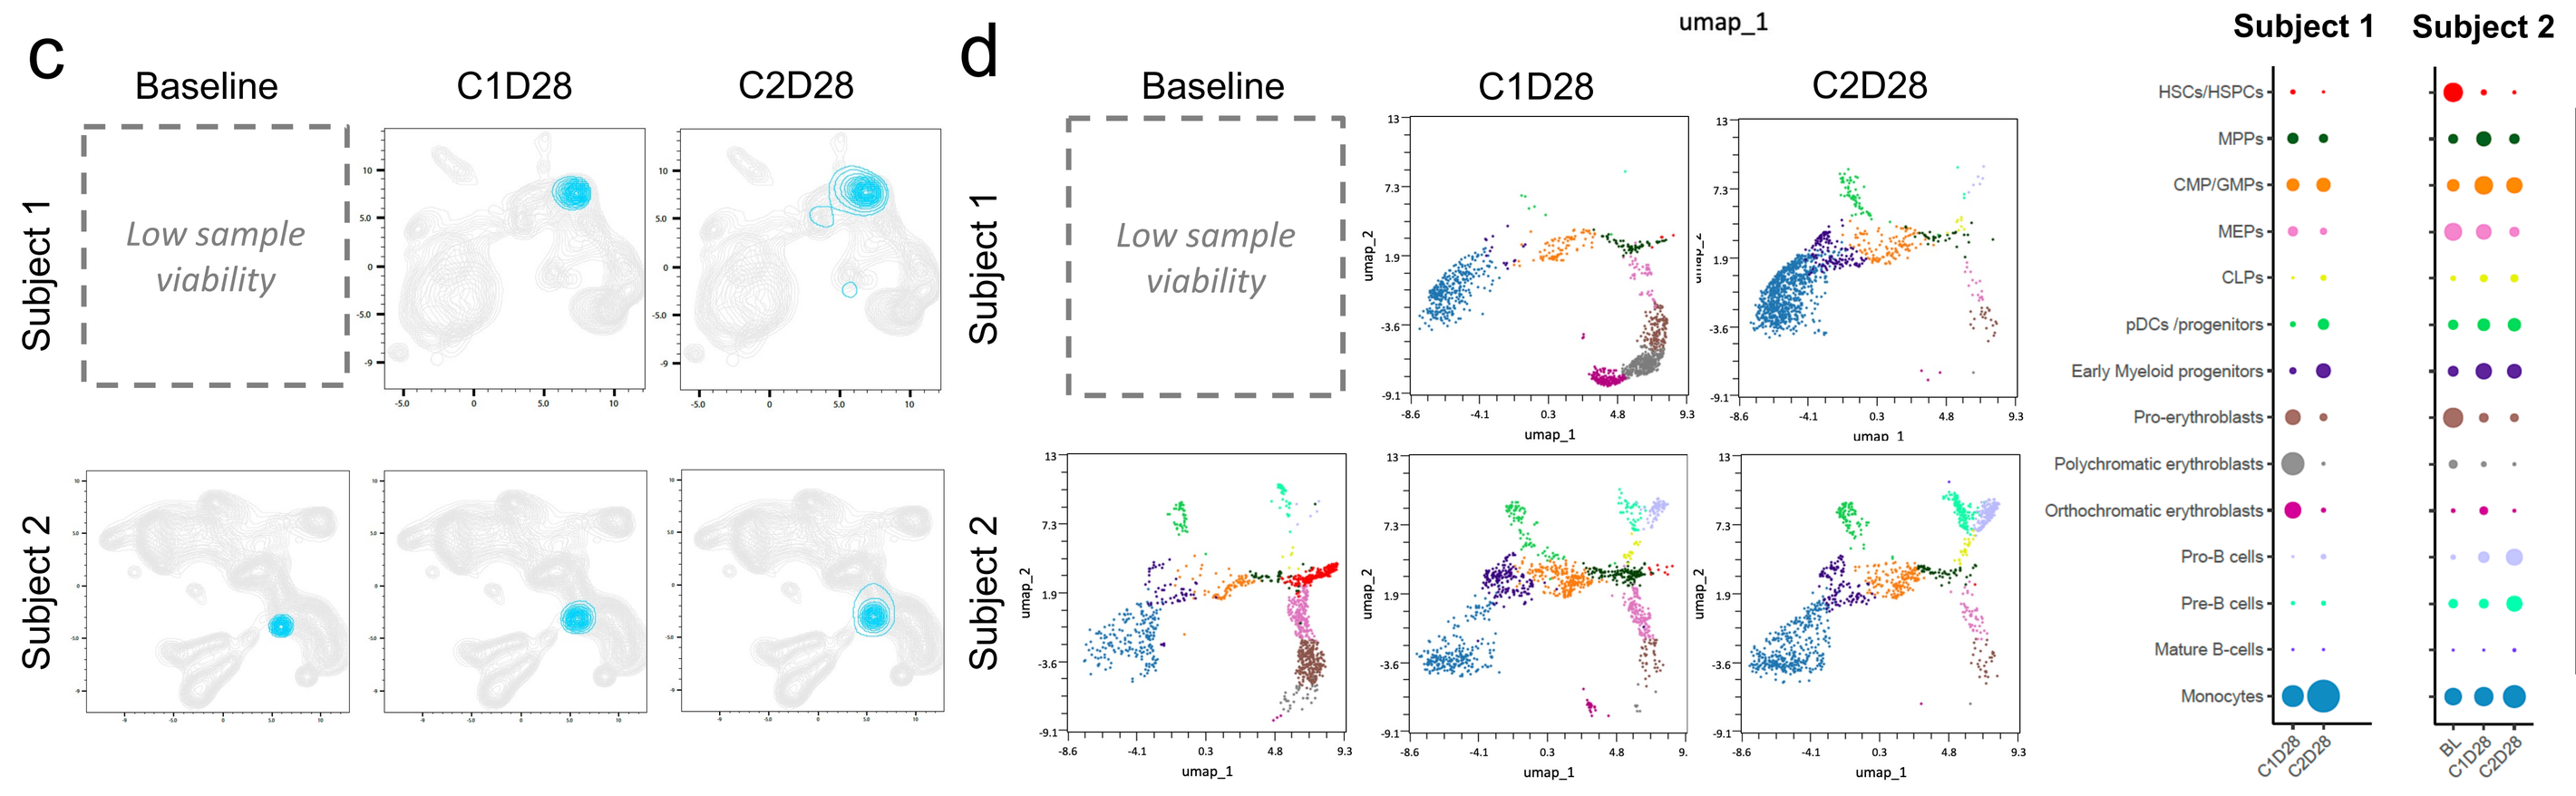
c
Baseline
C1D28
C2D28
Subject 1
Subject 2
Low sample viability
d
Baseline
C1D28
C2D28
Subject 1
Subject 2
Low sample viability
Subject 1
Subject 2

- Subjects achieving cCR retained HSPCs in their bone marrow.
- Observed preservation of hematopoietic trajectories in responders.
- Progenitor cells are enriched at baseline and preserved during SENTI-202 treatment.

Analysis of HSPCs and hematopoietic differentiation in the bone marrow of trial subjects. (a) HSPCs were identified as CD34+CD38-/low hematopoietic cells, and the proportion of HSPCs in responder bone marrow was either increased or maintained during SENTI-202 treatment. (b) CyTOF analysis identified cell populations in the classical hematopoietic differentiation hierarchy in healthy and diseased samples. Representative analysis was performed on responder subjects 1 and 2, showing that (c) EMCN+ hematopoietic populations (blue contours) were retained after SENTI-202 treatment. (d) UMAP analysis of subjects 1 and 2 showed the preservation of hematopoietic trajectories in responders, with progenitor cell types enriched at baseline and more differentiated populations appearing after treatment.

HSPC-Driven Immune Repopulation in Peripheral Blood after SENTI-202 Treatment

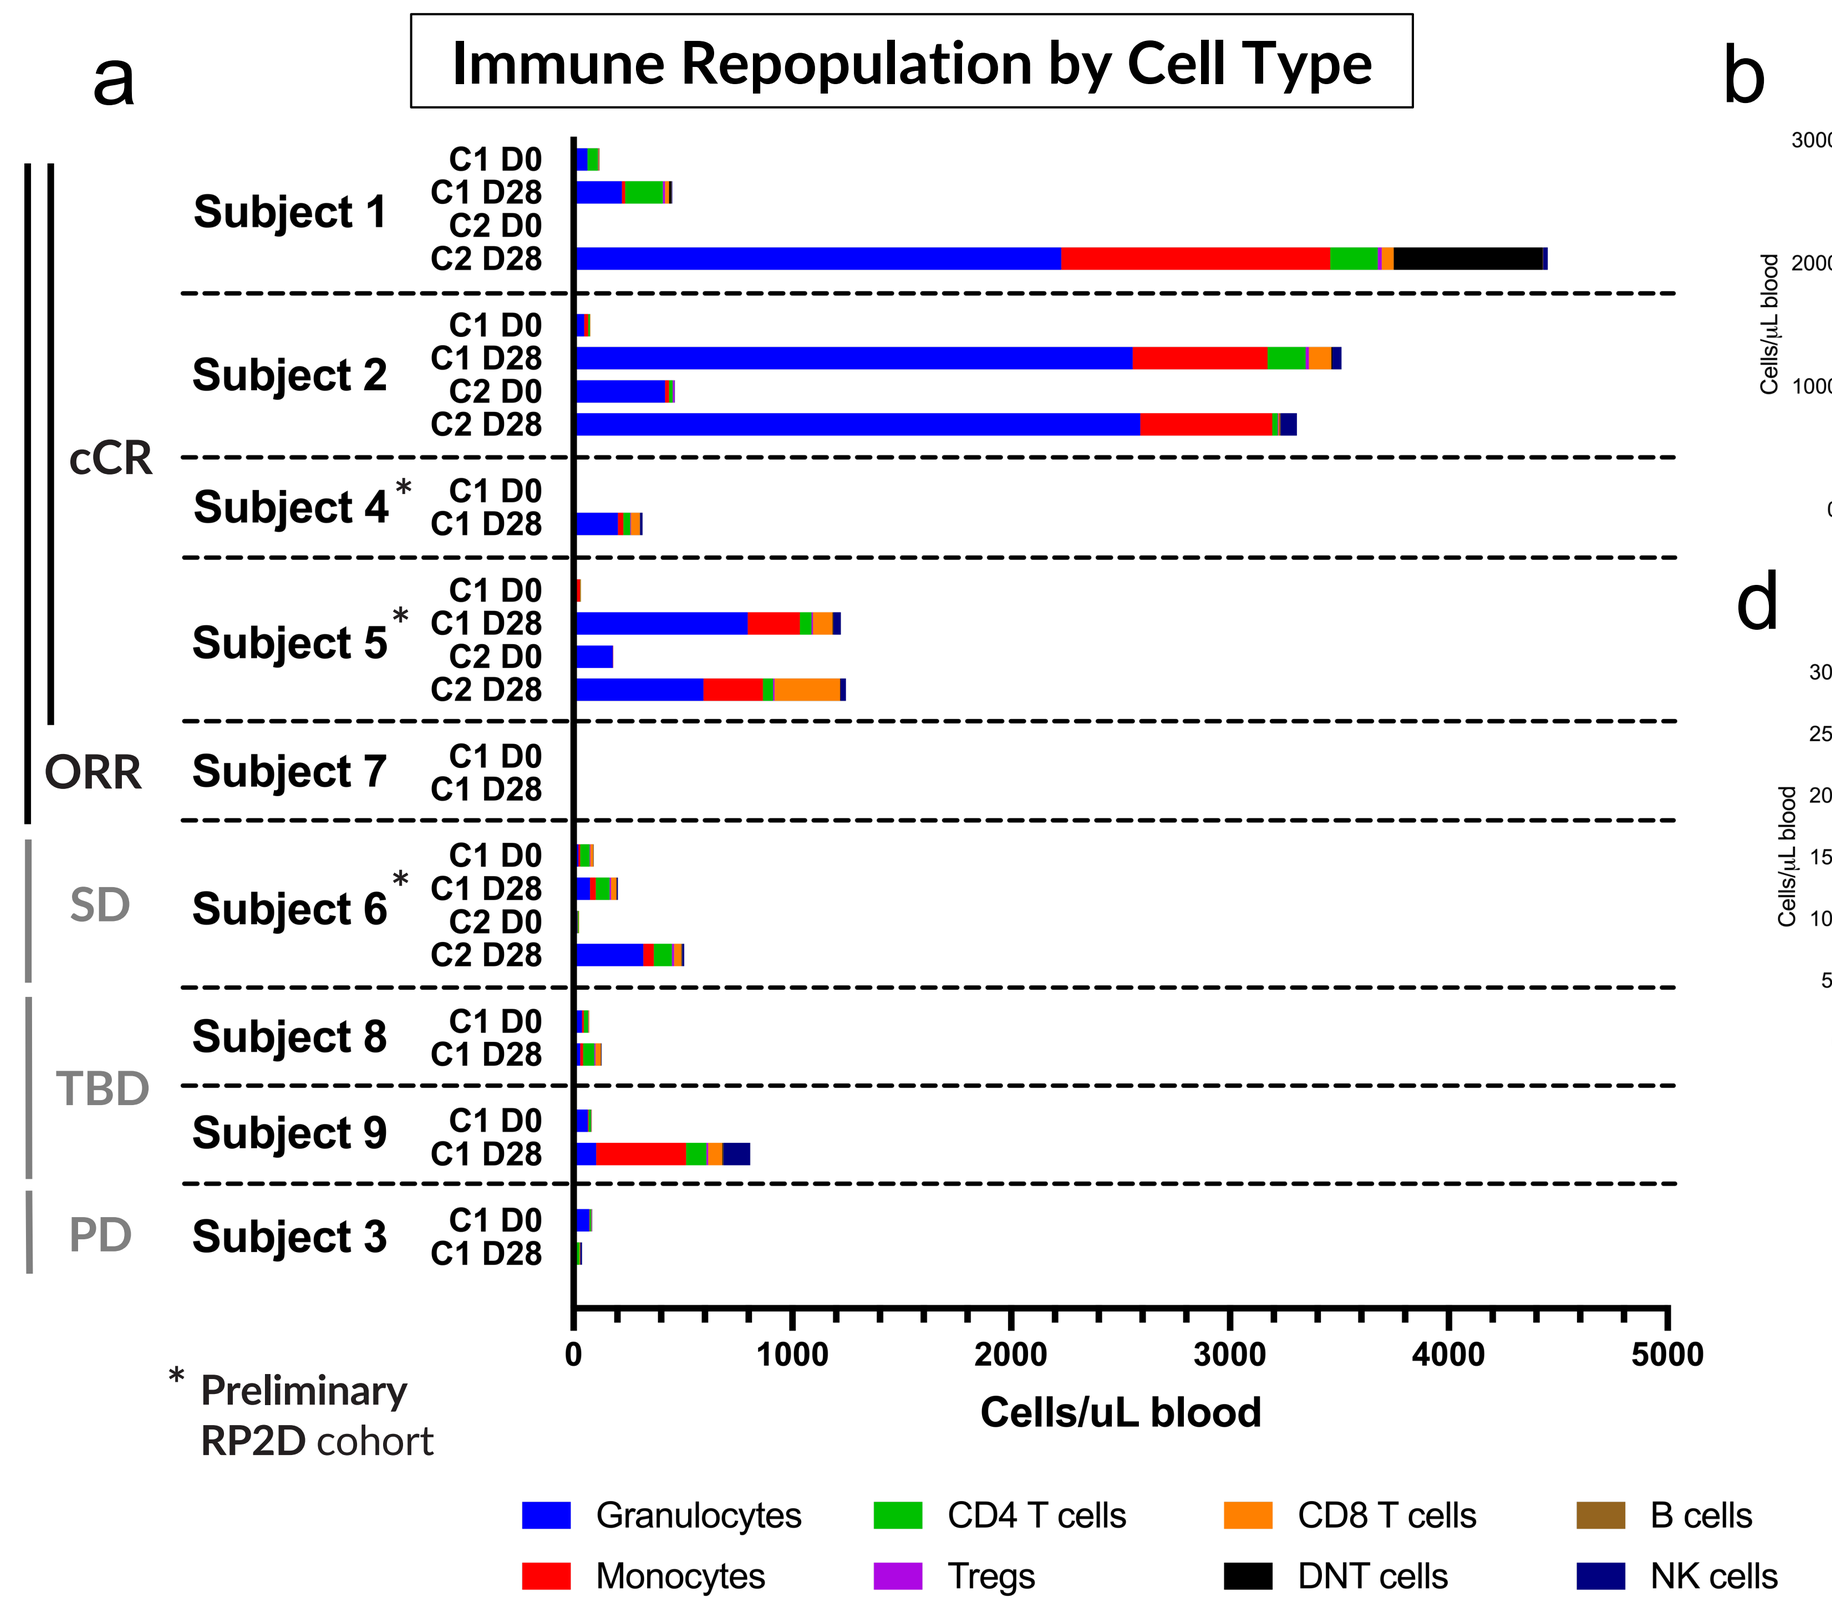
a
Immune Repopulation by Cell Type
cCR
ORR
SD
TBD
PD
Subject 1
Subject 2
Subject 4*
Subject 5*
Subject 7
Subject 6*
Subject 8
Subject 9
Subject 3
C1 D0
C1 D28
C2 D0
C2 D28
Cells/uL blood
* Preliminary RP2D cohort
Granulocytes
Monocytes
CD4 T cells
Tregs
CD8 T cells
DNT cells
B cells
NK cells

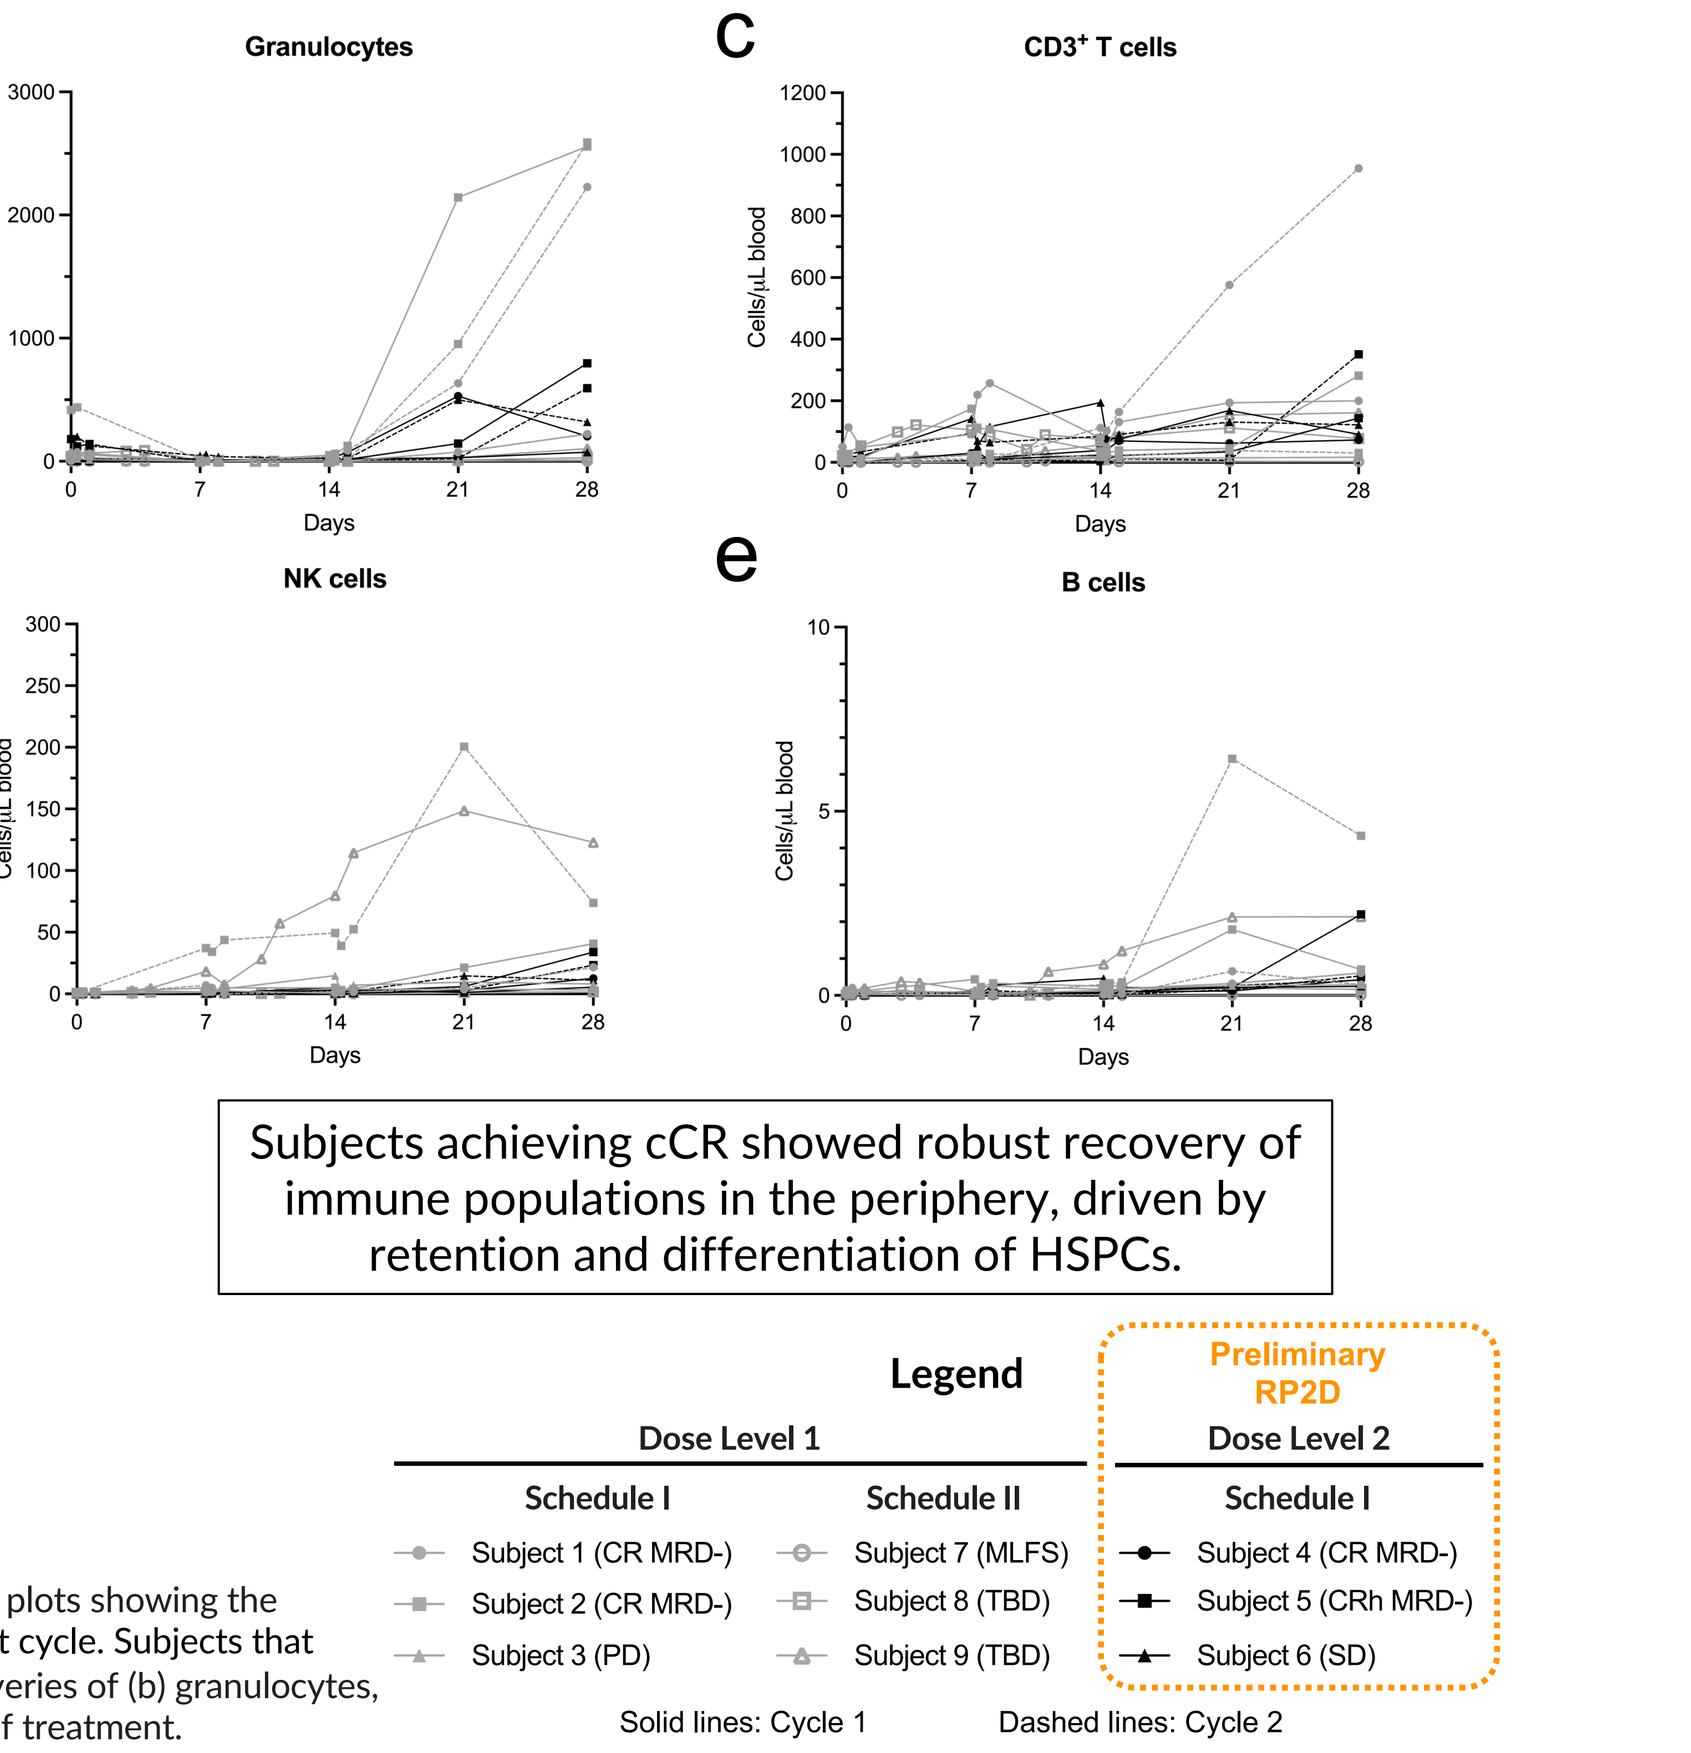
b
Granulocytes
c
CD3+ T cells
d
NK cells
e
B cells
Cells/μL blood
Days
Legend
Dose Level 1
Schedule I
Subject 1 (CR MRD-)
Subject 2 (CR MRD-)
Subject 3 (PD)
Schedule II
Subject 7 (MLFS)
Subject 8 (TBD)
Subject 9 (TBD)
Preliminary RP2D
Dose Level 2
Schedule I
Subject 4 (CR MRD-)
Subject 5 (CRh MRD-)
Subject 6 (SD)
Solid lines: Cycle 1
Dashed lines: Cycle 2

Subjects achieving cCR showed robust recovery of immune populations in the periphery, driven by retention and differentiation of HSPCs.

Analysis of immune populations in peripheral blood was performed by multicolor flow cytometry. (a) Stacked bar plots showing the concentration of cells measured in blood at the beginning (pre-dose 1) and end (Day 28 / EOT) of each treatment cycle. Subjects that achieved cCR showed robust immune recovery by the end of each cycle, indicating normal hematopoiesis. Recoveries of (b) granulocytes, (c) T cells, (d) NK cells, and (e) B cells are shown over each treatment cycle are shown for all subjects. EOT, end of treatment.

SUMMARY

- **AML blast reduction was observed in a majority of subjects.** 5/7 subjects evaluable for best overall response had blast reduction by CyTOF consistent with clinical response of CR or CRh (composite CR or cCR) or MLFS. 4/4 cCR patients were MRD- as assessed locally, 2/3 in preliminary RP2D cohort.
- **SENTI-202 was detected in all treated subjects**, with comparable PK to other CAR NK cell therapies.
- **AML in subjects enrolled on the SENTI-202-101 clinical trial was proteomically and mutationally heterogenous**, with a unique genetic and proteomic signature for each subject. All subjects expressed CD33 (range 28-99%), and 3/4 cCR subjects expressed FLT3 (range 5-46%). Most LSCs were in G0/G1 at baseline and were not expected to be eliminated by Flu/Ara-C.
- **In subjects achieving cCR:**
 - **A >10-fold decrease in LSCs was observed.**
 - **HSPCs were either increased or maintained after SENTI-202 treatment.** EMCN+ HSPCs were detectable throughout treatment and differentiated into normal hematopoietic lineages during response. **Multiple immune populations were increased in peripheral blood after SENTI-202 treatment.**
- Correlative data collected during the SENTI-202-101 clinical trial affirms the anti-leukemic effects of SENTI-202 in responders, as well as the repopulation of immune cell subpopulations post-treatment. The observed effects are consistent with the pharmacodynamic action of the Logic Gate in SENTI-202 in sparing EMCN+ HSPCs while selectively killing CD33+ and/or FLT3+ AML tumor cells.

ACKNOWLEDGEMENTS

The authors would like to thank all patients, their families, and caregivers for their participation in this study, as well as clinical and research staff at all participating institutions. Pharmacokinetics assessment was performed by Charles River Laboratories, and pharmacometrics support was provided by Certara.

Presented at The AACR Annual Meeting 2025, April 25 - 30, Chicago, IL

SENTI BIOSCIENCES, CONTACT
email: kanya.rajangam@sentibio.com